Abstract 9648

A Randomized, Double-Blind Phase 3 Trial of Extended-Release Granisetron (APF530) Versus Palonosetron for Preventing Chemotherapy-Induced Nausea and Vomiting Associated With Moderately or Highly Emetogenic Chemotherapy: Does a Reanalysis Using Newer ASCO Emetogenicity Criteria Affect Study Conclusions?

H. Raftopoulos,[1] R. Boccia,[2] W. Cooper,[3] E. O'Boyle,[4] R.J. Gralla[5]

[1]Hofstra North Shore-LIJ School of Medicine, Lake Success, NY; [2]Center for Cancer and Blood Disorders, Bethesda, MD; [3]TFS International, Flemington, NJ; [4]FibroGen, Inc., San Francisco, CA; [5]Albert Einstein College of Medicine, Bronx, NY

BACKGROUND

- The risk of chemotherapy-induced nausea and vomiting (CINV) is frequently reported as 31% to 90% with moderately emetogenic chemotherapy (MEC) and > 90% with highly emetogenic chemotherapy (HEC)
- Combination chemotherapeutic agents were classified by Hesketh et al according to their emetogenic potential in the absence of anetiemetic prophylaxis[1]
- In updated antiemesis guidelines, ASCO reclassified the emetogenicity of some chemotherapeutic regimens (eg, cyclophosphamide + anthracyclines [reclassified from MEC to HEC] and carboplatin-based regimens [reclassified from HEC to MEC])[2]
- Even with the latest generation of antiemetics, a need exists for improved prevention of CINV, especially delayed (24-120 h after chemotherapy)[3]
- APF530, a novel formulation of 2% granisetron and a bioerodible tri(ethylene glycol) poly(ortho ester) polymer, is designed to provide slow and sustained release of granisetron to prevent both acute (0-24 h after chemotherapy) and delayed CINV associated with MEC and HEC[4]
- In a phase 3 noninferiority trial (NCT00343460) comparing efficacy and safety of APF530 250 mg and 500 mg SC with the approved palonosetron dose (0.25 mg intravenous [IV]), both APF530 doses were noninferior to palonosetron in controlling acute CINV in patients receiving MEC or HEC; the higher dose was noninferior in preventing delayed CINV after MEC[5,6]
- Here, we present a post hoc analysis of efficacy data from the phase 3 trial; patients were reclassified as receiving MEC or HEC according to updated ASCO emetogenicity criteria, to establish whether the reclassification affected the original analysis findings[6]

METHODS

- Prospective, multicenter, randomized, double-blind, double-dummy, parallel-group phase 3 trial
- Adult (≥ 18 years old) men or women with histologically or cytologically confirmed malignancy scheduled to receive single-day MEC (Hesketh score 3 or 4) or HEC (Hesketh score 5)[1]
- Each patient was randomized to receive (**Figure 1**)
 - APF530 250 mg SC plus placebo IV or
 - APF530 500 mg SC plus placebo IV or
 - Palonosetron 0.25 mg IV plus placebo SC
- On completion of cycle 1 (C1), palonosetron was discontinued and consenting patients in that arm were rerandomized 1:1 to receive APF530 250 mg or 500 mg for C2-4
- Standard doses of IV dexamethasone (8 mg for MEC, 20 mg for HEC) were administered per protocol prior to chemotherapy on day 1; oral dexamethasone (8 mg bid) was given to HEC patients on days 2, 3, and 4. Rescue medication was permitted

Figure 1. Study Design

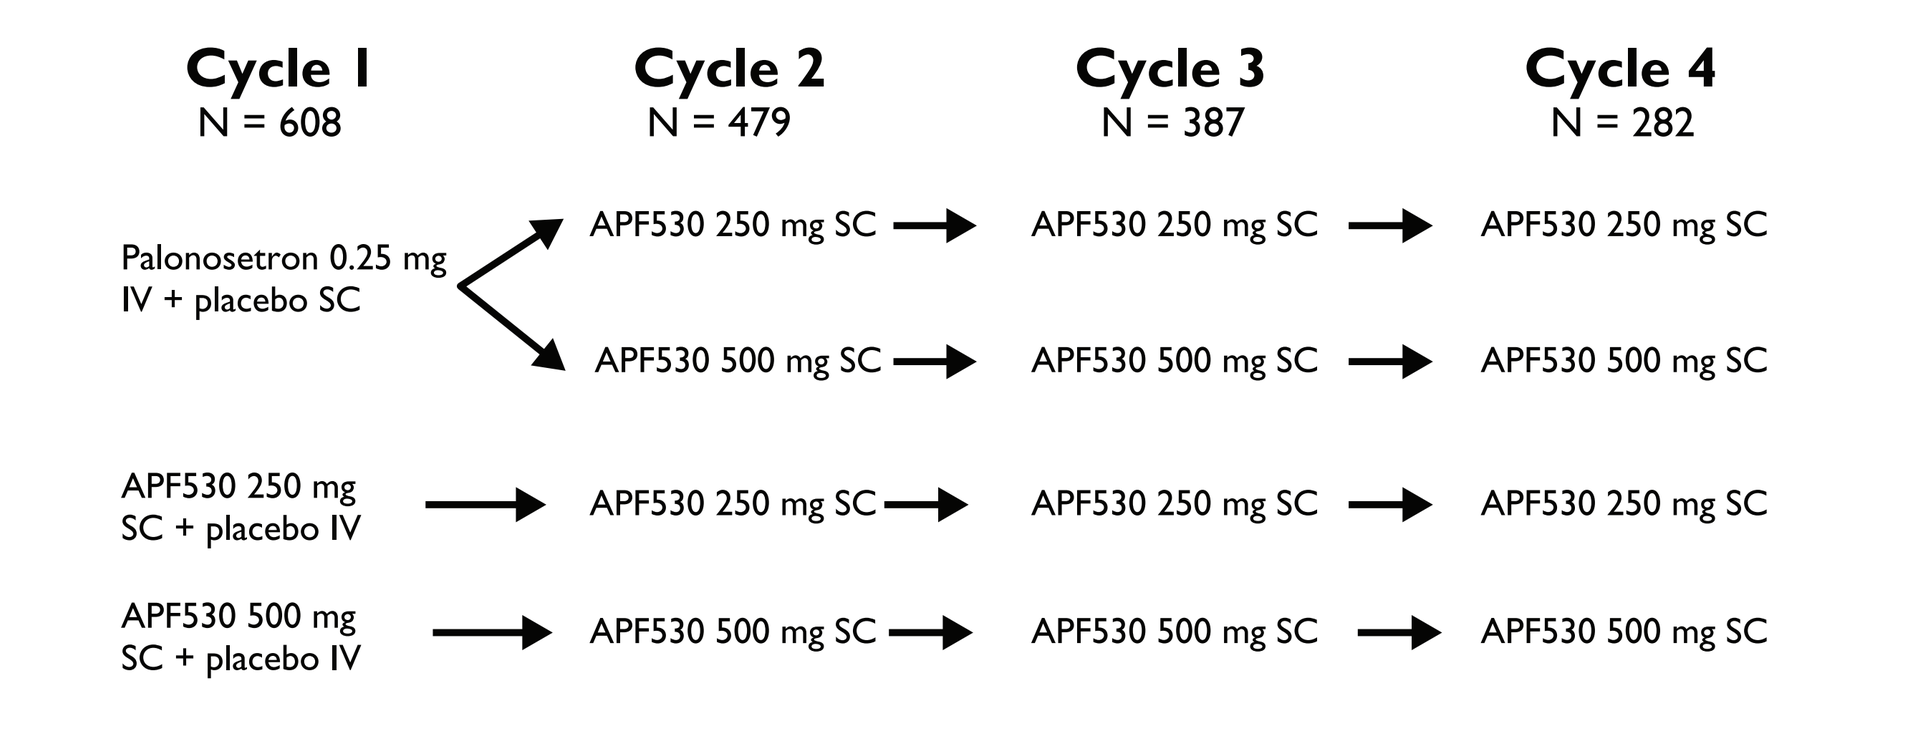
Cycle 1
N = 608
Cycle 2
N = 479
Cycle 3
N = 387
Cycle 4
N = 282
Palonosetron 0.25 mg IV + placebo SC
APF530 250 mg SC
APF530 250 mg SC
APF530 250 mg SC
APF530 500 mg SC
APF530 500 mg SC
APF530 500 mg SC
APF530 250 mg SC + placebo IV
APF530 250 mg SC
APF530 250 mg SC
APF530 250 mg SC
APF530 500 mg SC + placebo IV
APF530 500 mg SC
APF530 500 mg SC
APF530 500 mg SC

IV = intravenous; SC = subcutaneous.

- Primary objectives: establish noninferiority of APF530 for prevention of acute and delayed CINV after MEC or HEC versus palonosetron 0.25 mg IV, measured by complete response (CR; no emesis and no use of rescue medications) during C1
- Safety assessments: adverse events (AEs) during each treatment cycle, including type, duration, severity, and relation to study drug
- **For this post hoc analysis**
 - Regimens characterized by Hesketh criteria were reclassified as MEC or HEC using new ASCO emetogenicity guidelines
 - Efficacy comparisons between groups used Fisher's exact test
 - As with the original analysis, noninferiority was achieved if the lower bound of the confidence interval for the difference (APF530 - palonosetron) in CR rates was greater than -15%

RESULTS

Table 1. Patient Demographics and Baseline Clinical Characteristics (Safety Population)

	Moderately Emetogenic Chemotherapy			Highly Emetogenic Chemotherapy		
	APF530 250 mg n = 199	APF530 500 mg n = 216	Palo 0.25 mg n = 219	APF530 250 mg n = 256	APF530 500 mg n = 235	Palo 0.25 mg n = 225
Age, mean (SD), y	60.1 (12.6)	58.9 (13.3)	60.6 (12.8)	53.0 (12.6)	53.0 (12.0)	55.0 (12.6)
Female, n (%)	131 (65.8)	149 (69.0)	140 (63.9)	219 (85.5)	179 (76.2)	189 (84.0)
Race/ethnicity, n (%)						
White or Caucasian	124 (62.3)	137 (63.4)	147 (67.1)	136 (53.1)	135 (57.4)	134 (59.6)
Hesketh score, n (%)						
3	20 (10.1)	24 (11.1)	21 (9.6)	0	0	0
4	58 (29.1)	61 (28.2)	65 (29.7)	137 (53.5)	119 (50.6)	111 (49.3)
5	121 (60.8)	131 (60.6)	133 (60.7)	119 (46.5)	116 (49.4)	114 (50.7)
ECOG PS 0-1, n (%)	185 (93.0)	207 (95.8)	208 (95.0)	249 (97.3)	231 (98.3)	217 (96.4)
Received prior chemotherapy, n (%)	110 (57.0)	126 (61.5)	131 (62.1)	115 (47.7)	104 (45.0)	93 (42.7)

ECOG PS = Eastern Cooperative Oncology Group performance status; Palo = palonosetron.

- There were 1395 patients in the safety population, of whom 1350 (634 MEC, 716 HEC) were classified as at least MEC by ASCO criteria. There were 1299 (609 MEC, 690 HEC) patients in the modified intent-to-treat (mITT) population
- Demographics were similar in all treatment arms (**Table 1**)
- The most common tumor types were
 - MEC population: lung (32.2%), ovarian (22.8%), breast (19.4%)
 - HEC population: breast (68.8%), lung (6.2%), lymphoma (5.7%)
- The most common current chemotherapy regimens are shown in **Table 2**

Table 2. Most Common Current Chemotherapy Regimens (mITT)*

	Hesketh Score	APF530 250 mg	APF530 500 mg	Palonosetron 0.25 mg
MEC regimens, total n (%)		**n = 193**	**n = 205**	**n = 211**
Doxorubicin	3	8 (4.1)	8 (3.9)	10 (4.7)
Carboplatin	4	17 (8.8)	16 (7.8)	17 (8.1)
Cyclophosphamide-docetaxel	4	7 (3.6)	7 (3.4)	14 (6.6)
5-FU-cyclophosphamide-methotrexate	4	10 (5.2)	11 (5.4)	6 (2.8)
Carboplatin-paclitaxel	5	59 (30.6)	60 (29.3)	65 (30.8)
Carboplatin-docetaxel	5	17 (8.8)	25 (12.2)	21 (10.0)
Carboplatin-gemcitabine	5	18 (9.3)	14 (6.8)	14 (6.6)
HEC regimens, total n (%)		**n = 241**	**n = 231**	**n = 218**
Cyclophosphamide-doxorubicin	4	118 (49.0)	108 (46.8)	103 (47.2)
5-FU-cyclophosphamide-doxorubicin	5	19 (7.9)	19 (8.2)	19 (8.7)
5-FU-cyclophosphamide-epirubicin	5	10 (4.1)	14 (6.1)	14 (6.4)
Bleomycin-dacarbazine-doxorubicin-vinblastine	5	9 (3.7)	8 (3.5)	10 (4.6)
Cisplatin-gemcitabine	5	14 (5.8)	10 (4.3)	8 (3.7)
Cyclophosphamide-docetaxel-doxorubicin	5	15 (6.2)	10 (4.3)	7 (3.2)

*Received by 10 or more patients in any treatment group.
5-FU = 5-fluorouracil; mITT = modified intent-to-treat; MEC = moderately emetogenic chemotherapy; HEC = highly emetogenic chemotherapy.

- Complete response rates (**Figure 2, Table 3**)
 - CR rates with APF530 250 mg or 500 mg in C1 were not significantly different from those of palonosetron in preventing both acute and delayed emesis with MEC and HEC regimens
 - No notable differences were seen in CR in C1 for acute or delayed CINV during MEC or HEC between the ASCO analysis set and the original prespecified analysis set

Figure 2. Complete Response Comparisons for Overall Study Population and ASCO Reanalyzed Population in Cycle 1 (mITT)

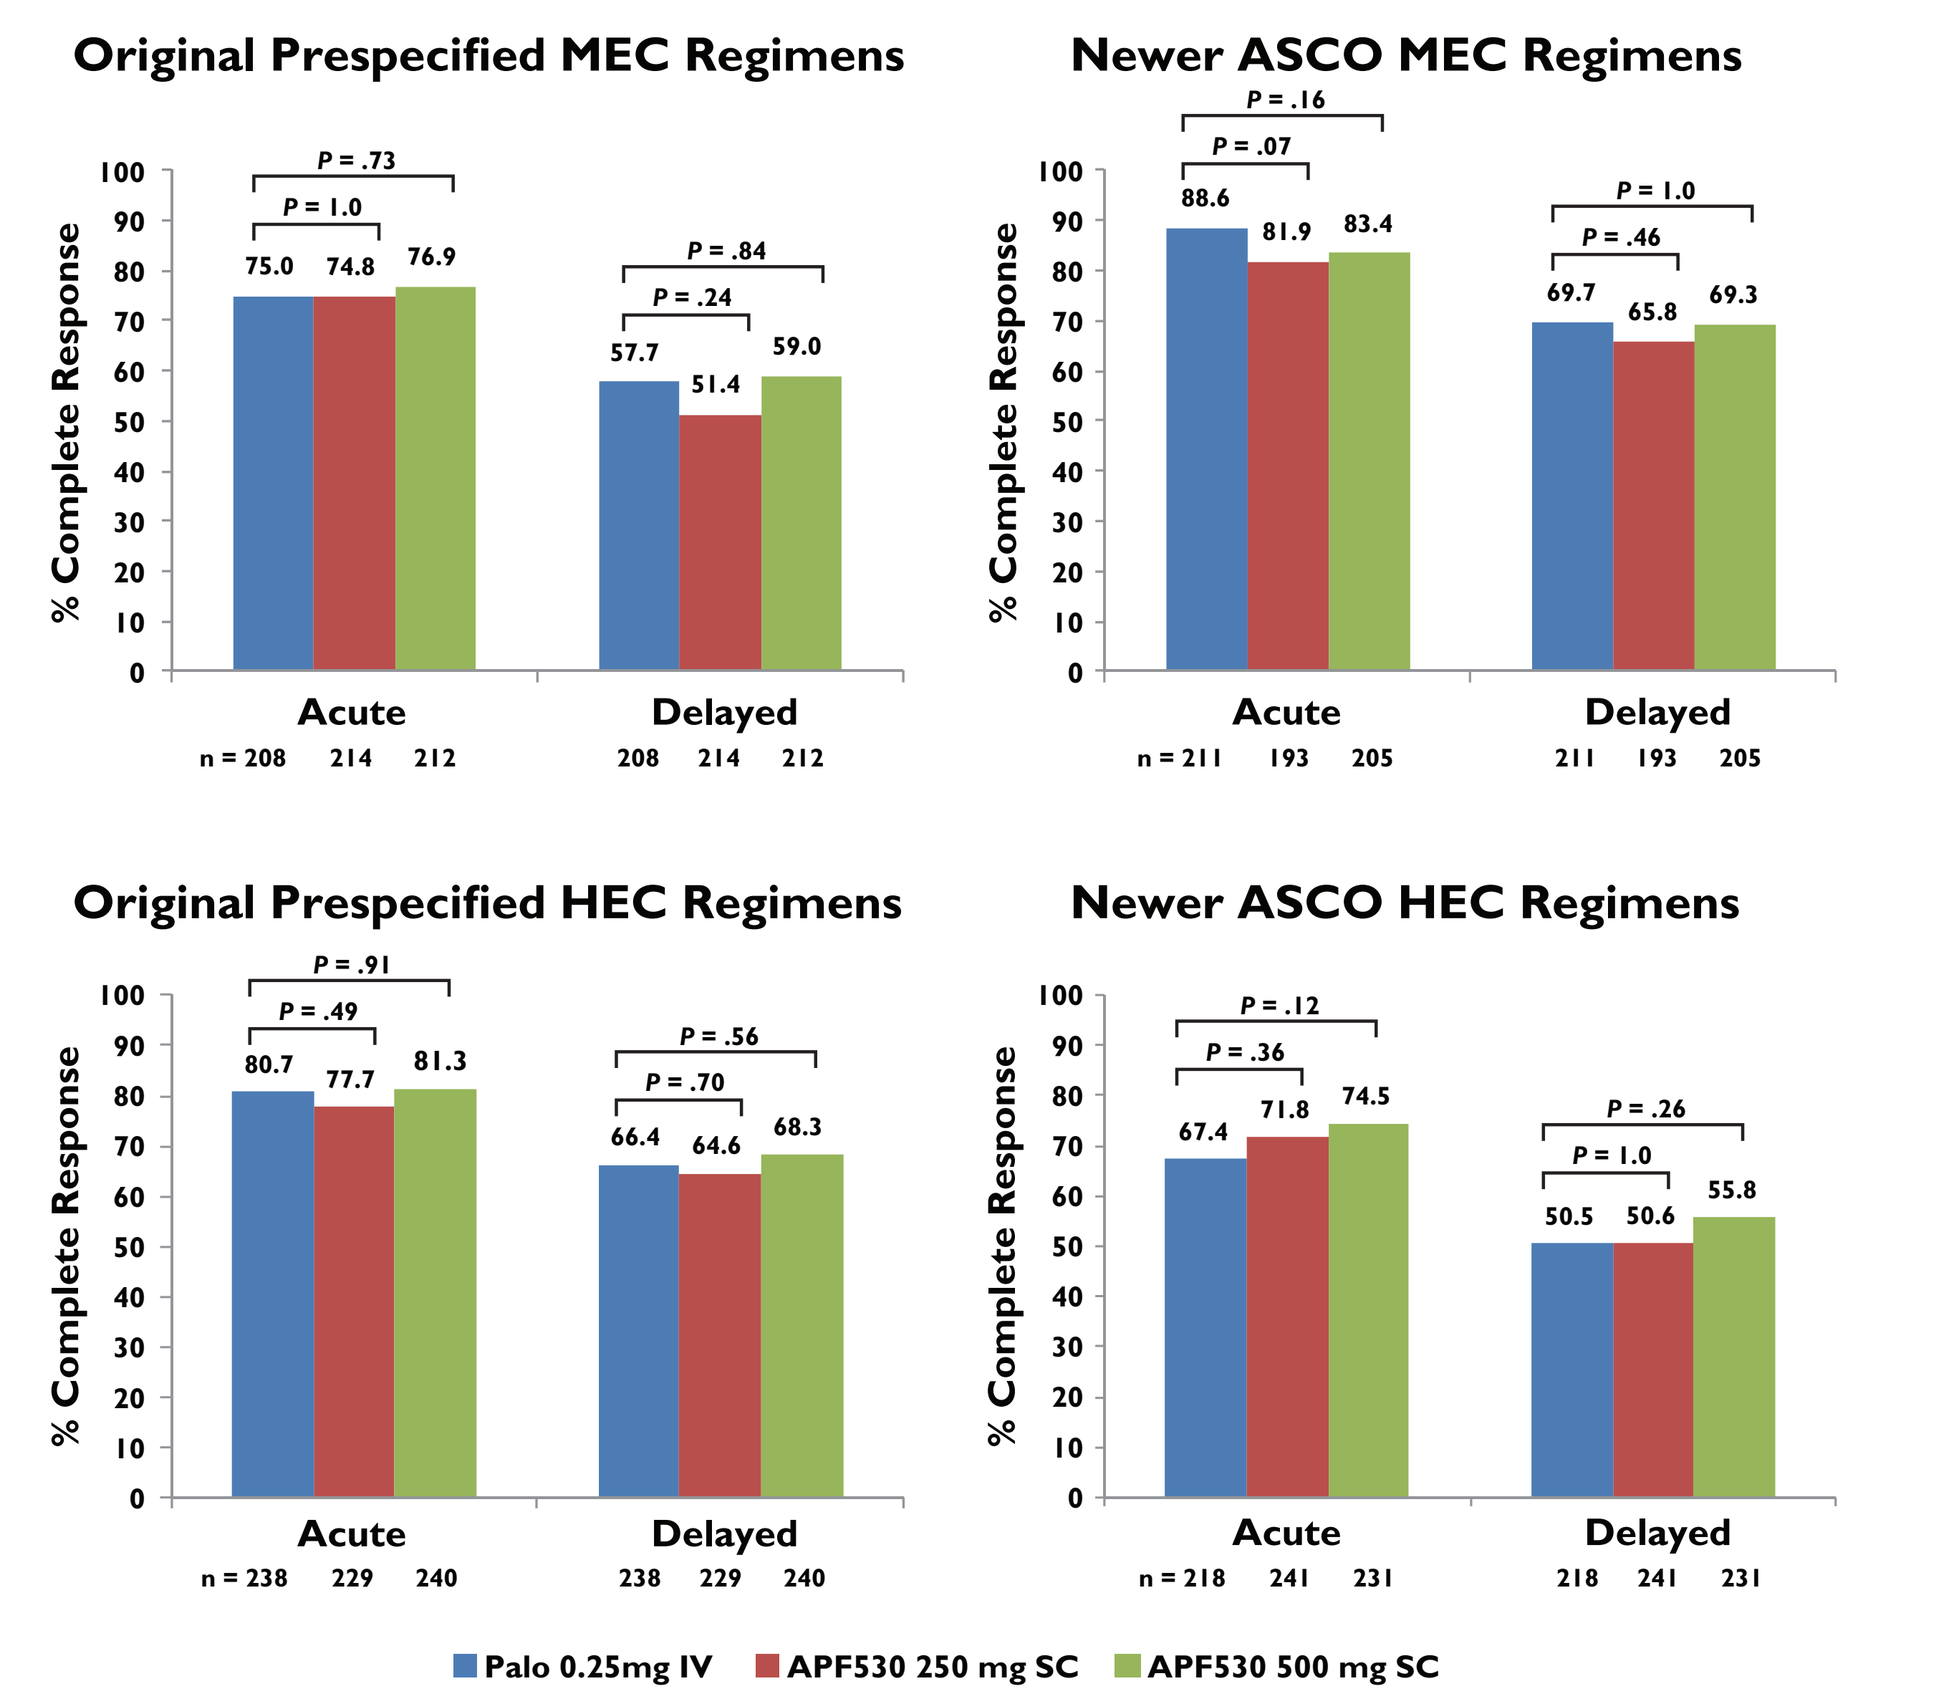
Original Prespecified MEC Regimens
% Complete Response
P = .73
P = 1.0
75.0
74.8
76.9
P = .84
P = .24
57.7
51.4
59.0
Acute
Delayed
n = 208 214 212
208 214 212
Newer ASCO MEC Regimens
P = .16
P = .07
88.6
81.9
83.4
P = 1.0
P = .46
69.7
65.8
69.3
n = 211 193 205
211 193 205
Original Prespecified HEC Regimens
P = .91
P = .49
80.7
77.7
81.3
P = .56
P = .70
66.4
64.6
68.3
n = 238 229 240
238 229 240
Newer ASCO HEC Regimens
P = .12
P = .36
67.4
71.8
74.5
P = .26
P = 1.0
50.5
50.6
55.8
n = 218 241 231
218 241 231
Palo 0.25mg IV
APF530 250 mg SC
APF530 500 mg SC

HEC = highly emetogenic chemotherapy; MEC = moderately emetogenic chemotherapy; mITT = modified intent-to-treat.

Table 3. Confidence Intervals (CI) for Difference (APF530 - Palonosetron) in Complete Response Rates

Chemotherapy Emetogenicity	CINV Phase	APF530 Dose, mg	95% CI	Conclusion
Prespecified criteria				
MEC	Acute	250	(-8.6-8.1)	Noninferior to palonosetron
		500	(-6.3-10.1)	Noninferior to palonosetron
	Delayed	250	(-15.3-3.7)	-
		500	(-8.2-10.8)	Noninferior to palonosetron
HEC*	Acute	250	(-10.4-4.5)	Noninferior to palonosetron
		500	(-6.5-7.7)	Noninferior to palonosetron
ASCO criteria				
MEC	Acute	250	(-14.0-0.2)	Noninferior to palonosetron
		500	(-12.1-1.5)	Noninferior to palonosetron
	Delayed	250	(-13.1-5.3)	Noninferior to palonosetron
		500	(-9.4-8.5)	Noninferior to palonosetron
HEC*	Acute	250	(-4.1-12.8)	Noninferior to palonosetron
		500	(-1.4-15.5)	Noninferior to palonosetron

*Noninferiority was not tested for complete response in the delayed phase for HEC regimens.
CINV = chemotherapy-induced nausea and vomiting; MEC = moderately emetogenic chemotherapy; HEC = highly emetogenic chemotherapy.

SAFETY

- The safety population comprised all patients who were randomized and received study drug (APF530 250 mg, n = 464; APF530 500 mg, n = 468; palonosetron 0.25 mg, n = 463) (**Table 4**)
- ~68% of patients in each group experienced an AE
- In C1, 1 patient in each APF530 treatment group discontinued because of an AE
- Excluding injection-site reactions (ISRs), the most common AEs across all groups were fatigue, constipation, and headache
- ISRs occurred across all treatment groups, and at higher rate in the APF530 groups relative to palonosetron; the most frequent events were bruising, erythema, and nodules
- After C1, 7 deaths occurred in the APF530 250 mg group, and 2 in each of the APF530 500 mg and palonosetron groups. No deaths were related to treatment
- 1 patient each in the APF530 250 mg and 500 mg groups discontinued because of a treatment-related AE in the HEC population (moderate dyspepsia for APF530 250 mg; mild drug hypersensitivity for APF530 500 mg)

Table 4. Treatment-Emergent Adverse Events (> 10%) in Any Group in Cycle 1 (Safety Population)

Adverse Events	APF530 250 mg n = 464	APF530 500 mg n = 468	Palonosetron 0.25 mg n = 463
Preferred term,* n (%)			
Constipation	62 (13.4)	72 (15.4)	62 (13.4)
Diarrhea	49 (10.6)	44 (9.4)	39 (8.4)
Fatigue	62 (13.4)	62 (13.2)	52 (11.2)
Headache	31 (6.7)	47 (10.0)	45 (9.7)
Injection-site reactions, n (%)			
Bruising	78 (16.8)	93 (19.9)	41 (8.9)
Erythema	33 (7.1)	51 (10.9)	14 (3.0)
Nodule	22 (4.7)	50 (10.7)	3 (0.6)

*Excludes hematologic adverse events (anemia, leukopenia, neutropenia), abdominal pain, alopecia, nausea, and vomiting, which were assumed to be related to chemotherapy.

CONCLUSIONS

- The original prespecified analysis of the phase 3 trial demonstrated noninferiority of APF530 to palonosetron in controlling acute CINV in patients receiving MEC or HEC; the higher dose was noninferior to palonosetron in preventing delayed CINV in patients receiving MEC[4-6]
- We then conducted this post hoc analysis to reanalyze the data according to the new ASCO emetogenicity criteria. There was no significant difference between APF530 and palonosetron in preventing acute and delayed CINV in patients receiving MEC or HEC, as determined by CR after C1
- Reclassifying chemotherapy emetogenicity by the new ASCO criteria did not alter study conclusions from the original prespecified analyses regarding noninferiority to palonosetron in acute and delayed MEC and acute HEC (which was established) or superiority to palonosetron in delayed HEC (which was not established)
 - However, reclassifying chemotherapy emetogenicity by the new ASCO criteria resulted in better CR rates in the MEC groups and poorer CR rates in the HEC groups for all arms of the study. This supports the ASCO reclassification
- Single-dose APF530 SC is an effective alternative to palonosetron for preventing acute and delayed CINV after MEC or HEC, with generally mild and manageable AEs

REFERENCES

1. Hesketh et al. *J Clin Oncol*. 1997;15:103-109.
2. Basch et al. *J Clin Oncol*. 2011;29:4189-4198.
3. Roila et al. *Ann Oncol*. 2010;21(suppl 5):v232-v243.
4. Grous et al. ASCO 2009. Abstract 9627.
5. Boccia et al. ASCO 2013. Abstract 9626.
6. Charu et al. MASCC 2012. Abstract 1109.

Research support was provided by Heron Therapeutics, Inc. (formerly A.P. Pharma, Inc.)

Copies of this poster obtained through Quick Response Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

This presentation is the intellectual property of the author/presenter. Contact at harry.raftopoulos@gmail.com for permission to reprint and/or distribute.