Long-Term Safety and Efficacy of Lubiprostone for the Treatment of Chronic Idiopathic Constipation

John Johanson, MD* Raymond Panas, PhD** Aimee Wahle, MS** Ryuji Ueno, MD, PhD, PhD**
* University of Illinois College of Medicine, Rockford, Illinois, US; ** Sucampo Pharmaceuticals, Inc., Bethesda, Maryland, US

Abstract

Constipation is a common gastrointestinal condition with often ineffective treatment options. Lubiprostone, a type-2 chloride channel activator approved in the US for treatment of adult constipation and irritable bowel syndrome with constipation in adult women, has been shown to be efficacious and well tolerated by people with chronic constipation. Results are presented here from three open-labeled, long-term [6 to 12 months] trials where assessments were captured approximately every 6 weeks. Methods: Study 1 was a 24-week, open-labeled extension to a 4-week double-blinded, pivotal trial that enrolled 308 subjects. Study 2 was a 48-week, open-labeled trial in 250 subjects, 82 of whom participated in a 7-week randomized withdrawal study period prior to the open-labeled phase of the study. Study 3 was a 48-week open-labeled trial in 325 treatment-naïve subjects. Subjects assessed treatment effectiveness, constipation severity, and abdominal symptoms of bloating and discomfort using a 5-point scale. Results: Improvements in constipation severity, abdominal bloating, and abdominal discomfort were statistically significant at all visits (p<0.0001). Constipation severity was improved by an average of 1.28 points at Weeks 4 to 6 (N=828), 1.47 points at Week 24 (N=512), 1.38 points at Week 48 (Study 2 and 3 only; N=281), and 1.15 points for the last on-drug measurement (N=866). Abdominal bloating was improved by an average of 0.89 point at Weeks 4 to 6 (N=829), 0.98 point at Week 24 (N=512), 1.00 point at Week 48 (Study 2 and 3 only, N=282), and 0.79 point for the last on-drug measurement (N=867). Abdominal discomfort was improved by an average of 0.85 point at Weeks 4 to 6 (N=829), 0.91 point at Week 24 (N=512), 0.87 point at Week 48 (Study 2 and 3 only, N=282), and 0.72 point for the last on-drug measurement (N=867). Lubiprostone was well tolerated. The most commonly reported related adverse events (≥5% of subjects) were nausea, diarrhea, headache, abdominal distension, flatulence, and abdominal pain. Conclusion: Clinical improvements observed in prior short-term, double-blinded trials are maintained for at least 24 to 48 weeks, as shown in these three long-term trials. Lubiprostone provides significant relief for a variety of constipation symptoms, and efficacy is sustained for up to 48 weeks.

Introduction

- Approximately 5% of the US population, or nearly 15 million people, suffer from chronic idiopathic constipation.[1]
- Overall, approximately 15% of the population meets the Rome II criteria for constipation.[2]
- Lubiprostone selectively activates type-2 chloride channels (ClC-2) and enhances fluid secretion into the intestinal lumen.[3,4]
- Lubiprostone is approved in the United States for the treatment of chronic idiopathic constipation in adults, regardless of gender.[5]
- We present here the pooled data from 6- and 12-month long-term safety and efficacy studies of lubiprostone in subjects with chronic idiopathic constipation.

Methods

Entry Criteria

- Chronic constipation is defined as <3 spontaneous bowel movements (SBMs)/week, with a minimum 3-month history of hard stools, sensation of incomplete evacuation, or straining during at least 25% of bowel movements (BMs).
 - A SBM was any BM that did not occur within 24 hours of rescue medication use.
- Subjects were excluded for the following:
 - Documented mechanical obstruction;
 - Organic disorders of the bowel;
 - Constipation secondary to a documented cause (e.g., surgery, bowel resection);
 - Clinically significant cardiovascular, liver, lung, neurologic, or psychiatric disorder;
 - Clinically significant laboratory abnormalities.
- Some subjects in the 6-month study were follow-on subjects from a previous 4-week pivotal safety and efficacy study of lubiprostone.

Dose Administration

- After a 15-day drug-free period, eligible subjects received oral lubiprostone 24 mcg twice daily (BID) to be taken with food and water for 6 to 12 months, as needed.
 - "Subject need" was defined as perceived severity of constipation and need for relief.
- Subjects could then remain on a daily dosing schedule or stop the study drug if the perceived need decreased or ceased; subjects could return to study drug when needed, but were to restart dosing again of lubiprostone at 24 mcg BID.
- Investigators could also adjust the daily dose in response to exaggerated pharmacodynamic events (e.g., diarrhea) or treatment-related adverse events (AEs)—e.g., nausea.
- Periodic use of rescue medication, at the discretion of the investigator, was also permitted.

Safety and Efficacy Evaluations

- Evaluations were performed at baseline and at 4-week and 6-week intervals during treatment in the 6-month and 12-month studies. Follow-up evaluations were also performed 1 week or 2 weeks after completion of dose administration.
- Safety was assessed by AE incidence rates and efficacy was evaluated by improvements in subject assessments of constipation severity, abdominal bloating, and abdominal discomfort.
- All efficacy assessments were based on five point severity scales, where 0=Absent, 1=Mild, 2=Moderate, 3=Severe, and 4=Very Severe.
- Efficacy assessment data are summarized for the intent-to-treat (ITT) population, which includes all subjects who took at least one dose of study medication and completed one post-baseline efficacy assessment.

Results

Eight-hundred seventy eight (878) subjects were enrolled and treated with lubiprostone 48 mcg daily (24 mcg BID). Table 1 presents the demographic information for subjects included in the ITT population (n=871). Approximately 300 subjects participated in the 6-month and each of the two 12-month studies.

Table 1. Demographics (ITT population)

		Lubiprostone 24 mcg BID *N=871* *n (%)*
Mean Age in Years (range)		51 (19.0-86.0)
Gender	**Male**	121 (13.9)
	Female	750 (86.1)
Race	**Caucasian**	757 (86.9)
	African-American	64 (7.3)
	Asian	39 (4.5)
	Hispanic	6 (0.7)
	Other	5 (0.6)

E001734

Safety

The most commonly reported related adverse events (≥5% of subjects) were nausea, diarrhea, headache, abdominal distension, flatulence, and abdominal pain. No subjects died during the studies. Overall, 31 subjects reported a total of 44 serious adverse events (SAEs). Two SAEs were assessed by the investigator as possibly treatment-related: bilateral clubfoot and diarrhea. It should be noted that one female subject became pregnant during the studies and was immediately discontinued from treatment. Subsequently, a healthy baby was delivered with bilateral clubfoot.

Table 2 presents the AEs that were considered at least "possibly" related to study drug and that occurred in at least 2% of all subjects.

Table 2. Summary of Most Common (≥2%) Related* AEs (Safety-evaluable [SE]† Population)

	Lubiprostone 24 mcg BID *N=878 n (%)*
At Least 1 Event	480 (54.7)
Gastrointestinal Disorders	418 (47.6)
Nausea	224 (25.5)
Diarrhea	108 (12.3)
Abdominal Distension	59 (6.7)
Flatulence	47 (5.4)
Abdominal Pain	44 (5.0)
Loose Stools	32 (3.6)
Vomiting	29 (3.3)
Nervous System Disorders	105 (12.0)
Headache	85 (9.7)
Dizziness	23 (2.6)

*Includes all events with a relationship rating of "Possible," "Probable," or "Definite." †Safety-evaluable population includes those subjects who took at least one dose of study medication.

Efficacy

With regard to long-term efficacy, improvements in assessments of constipation severity, abdominal bloating, and abdominal discomfort were all statistically significant at all post-baseline timepoints in all studies and at all visits (p<0.0001).

As shown in Figure 1, mean improvements from baseline in ratings of constipation severity ranged from 0.94 to 1.7 across the studies.

Figure 1. Subjective Assessments of Constipation Severity With Long-term Lubiprostone Treatment (ITT Population)

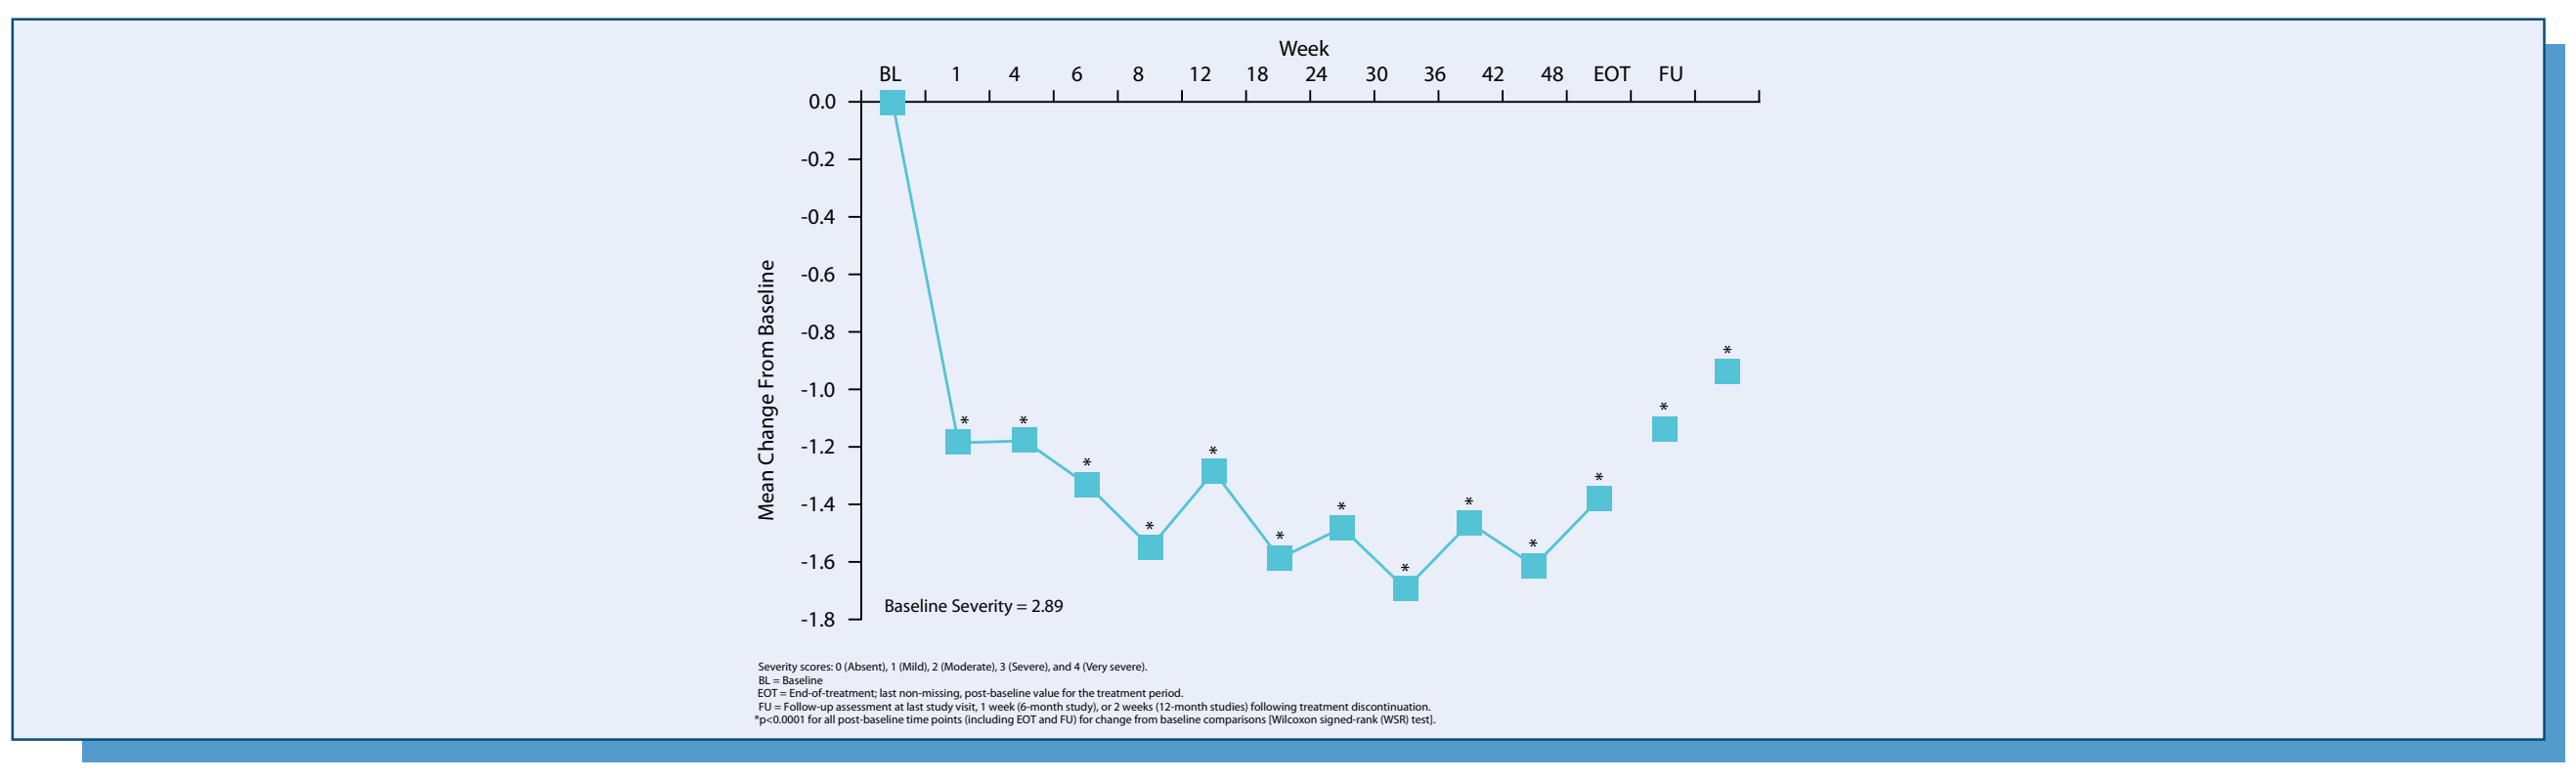
Week
BL 1 4 6 8 12 18 24 30 36 42 48 EOT FU
0.0 -0.2 -0.4 -0.6 -0.8 -1.0 -1.2 -1.4 -1.6 -1.8
Mean Change From Baseline
Baseline Severity = 2.89
Severity scores: 0 (Absent), 1 (Mild), 2 (Moderate), 3 (Severe), and 4 (Very severe).
BL = Baseline
EOT = End-of-treatment; last non-missing, post-baseline value for the treatment period.
FU = Follow-up assessment at last study visit, 1 week (6-month study), or 2 weeks (12-month studies) following treatment discontinuation.
*p<0.0001 for all post-baseline time points (including EOT and FU) for change from baseline comparisons [Wilcoxon signed-rank (WSR) test].

As shown in Figure 2, mean improvements in abdominal bloating ranged from 0.71 to 1.18 points across the studies.

Figure 2. Subjective Assessments of Abdominal Bloating With Long-term Lubiprostone Treatment (ITT Population)

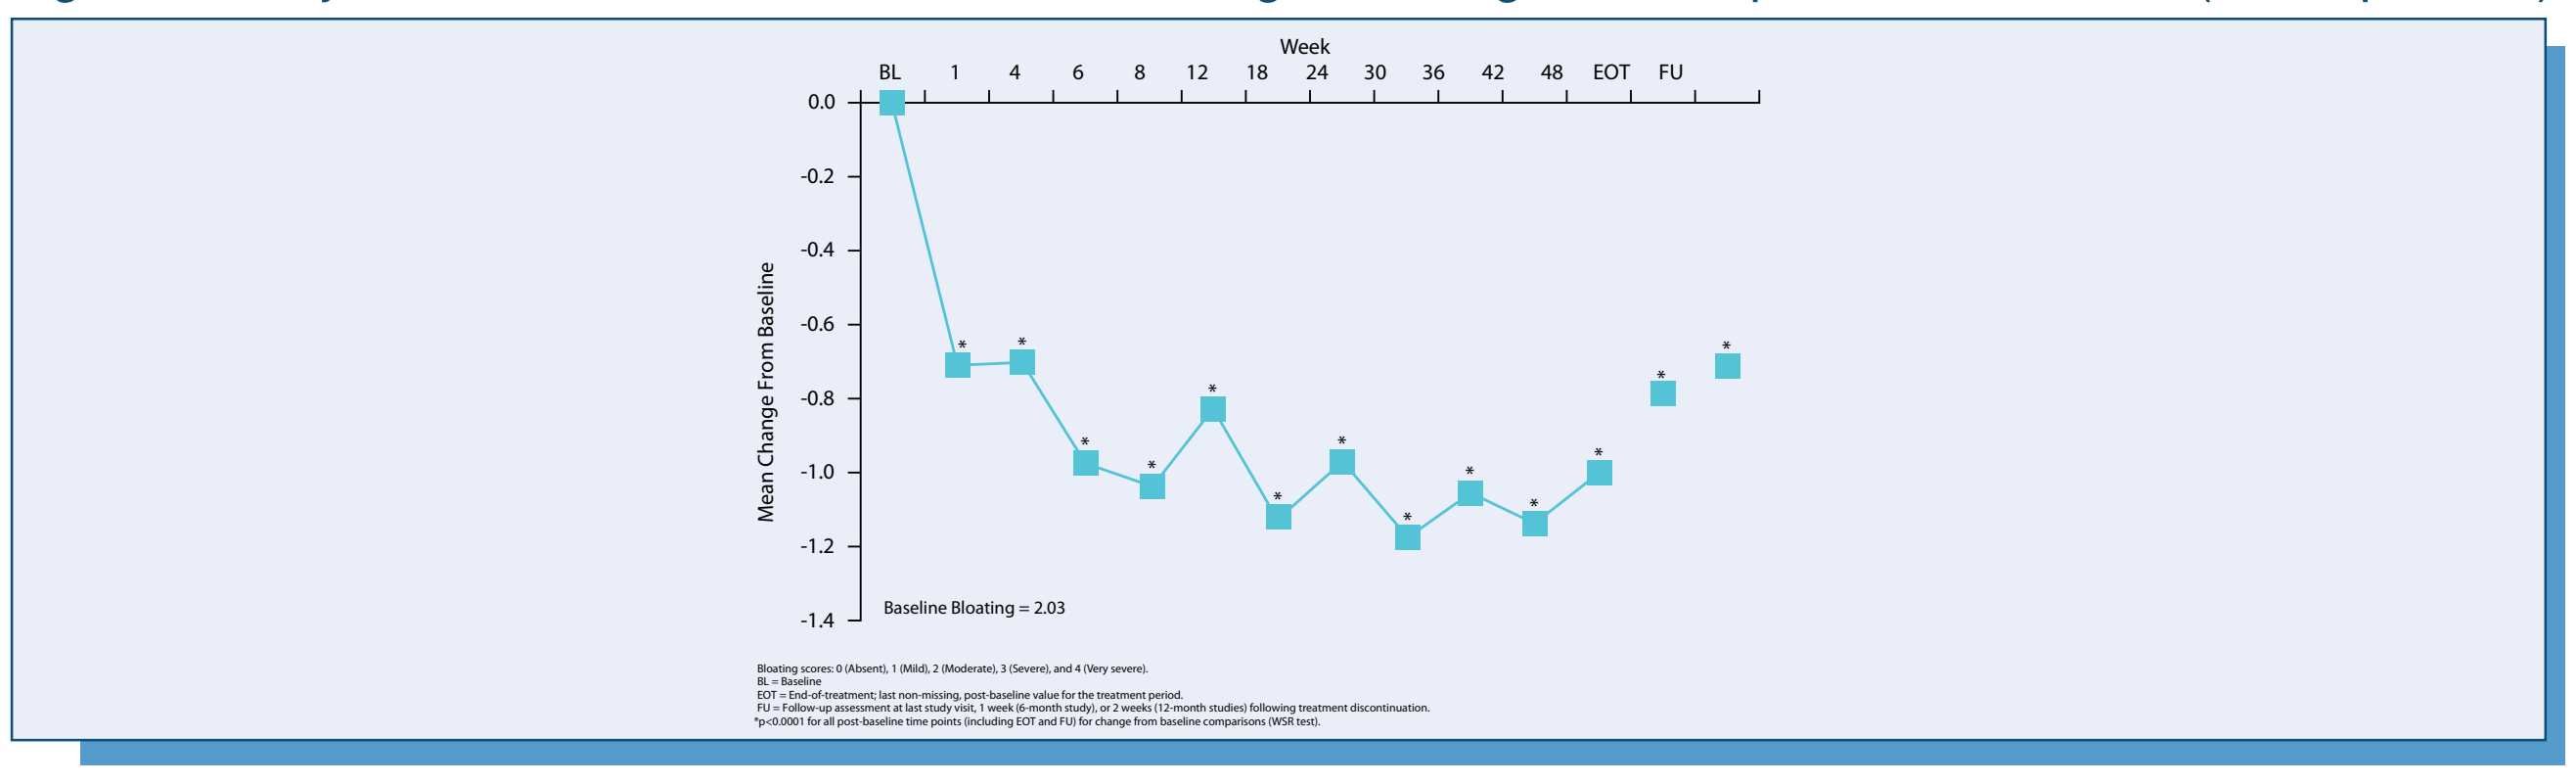
Week
BL 1 4 6 8 12 18 24 30 36 42 48 EOT FU
0.0 -0.2 -0.4 -0.6 -0.8 -1.0 -1.2 -1.4
Mean Change From Baseline
Baseline Bloating = 2.03
Bloating scores: 0 (Absent), 1 (Mild), 2 (Moderate), 3 (Severe), and 4 (Very severe).
BL = Baseline
EOT = End-of-treatment; last non-missing, post-baseline value for the treatment period.
FU = Follow-up assessment at last study visit, 1 week (6-month study), or 2 weeks (12-month studies) following treatment discontinuation.
*p<0.0001 for all post-baseline time points (including EOT and FU) for change from baseline comparisons (WSR test).

As shown in Figure 3, mean improvements in abdominal discomfort in subjects ranged from 0.66 to 1.07 points across the studies.

Figure 3. Subjective Assessments of Abdominal Discomfort With Long-term Lubiprostone Treatment (ITT Population)

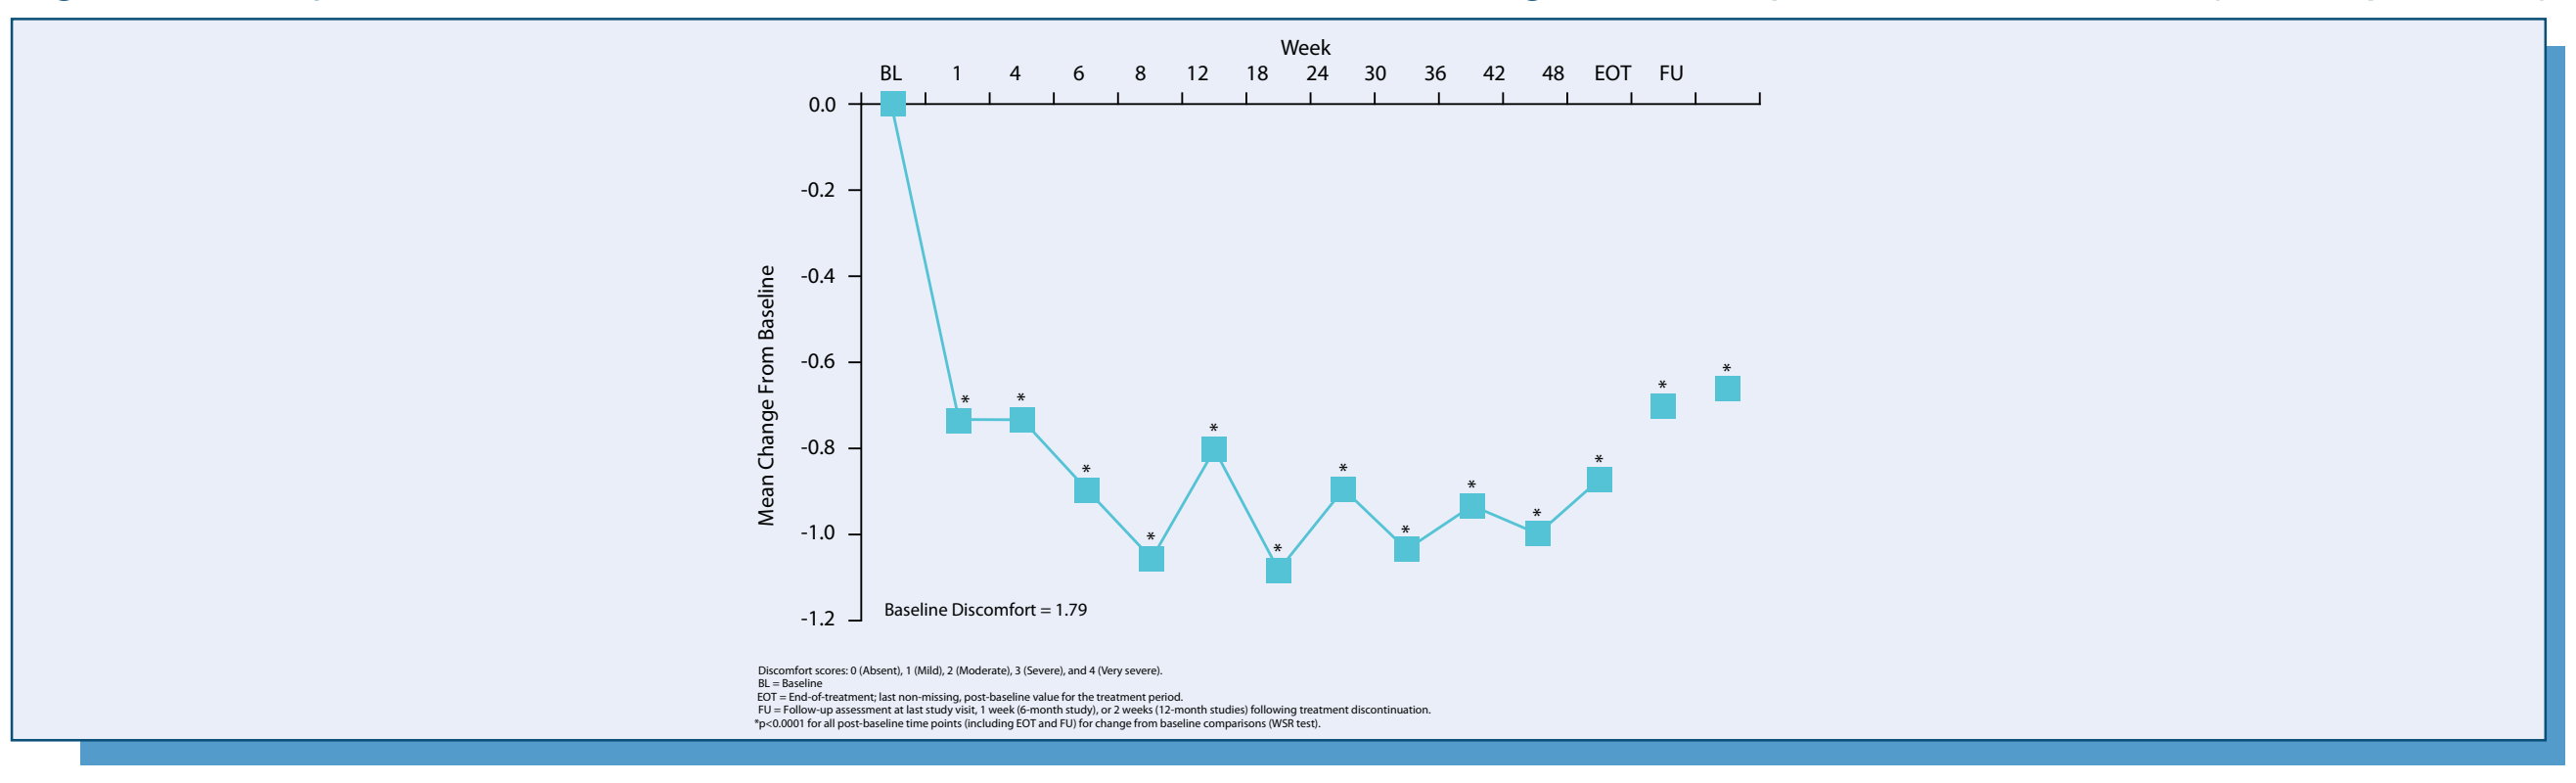
Week
BL 1 4 6 8 12 18 24 30 36 42 48 EOT FU
0.0 -0.2 -0.4 -0.6 -0.8 -1.0 -1.2
Mean Change From Baseline
Baseline Discomfort = 1.79
Discomfort scores: 0 (Absent), 1 (Mild), 2 (Moderate), 3 (Severe), and 4 (Very severe).
BL = Baseline
EOT = End-of-treatment; last non-missing, post-baseline value for the treatment period.
FU = Follow-up assessment at last study visit, 1 week (6-month study), or 2 weeks (12-month studies) following treatment discontinuation.
*p<0.0001 for all post-baseline time points (including EOT and FU) for change from baseline comparisons (WSR test).

Summary and Conclusion

- In subjects treated for 6 and 12 months with lubiprostone 48 mcg total daily dose (24 mcg BID), significant improvements from baseline were seen across all weeks for:
 - Constipation severity
 - Abdominal bloating
 - Abdominal discomfort
- Follow-up assessments of efficacy also showed significant improvements over baseline assessments.
- Overall, lubiprostone was well-tolerated. The most commonly-reported, related AEs (≥5% of subjects) were nausea, diarrhea, headache, abdominal distension, flatulence, and abdominal pain.
- Lubiprostone 48 mcg (24 mcg BID) provides sustained relief of symptoms over long-term treatment periods (6 and 12 months) in subjects with chronic idiopathic constipation.

References

1. Stewart WF, Liberman JN, Sandler RS, et al. Epidemiology of constipation (EPOC) study in the United States: relation of clinical subtypes to sociodemographic features. *Am J Gastroenterol.*1999;94:3530-3540.
2. Higgins PD, Johanson JF. Epidemiology of constipation in North America: a systematic review. *Am J Gastroenterol*. 2004;99:750-759.
3. Cuppoletti J, Malinowska DH, Tewari KP, et al. SPI-0211 activates T84 cell chloride transport and recombinant human ClC-2 chloride currents. *Am J Physiol Cell Physiol*. 2004;287:C1173-C1183.
4. Ueno R, Osama H, Habe T, Engelke K, Patchen M. Abstract #M1109. Oral SPI-0211 increases intestinal fluid secretion and chloride concentration without altering serum electrolyte levels. *Gastroenterology*. 2004;126(4 Suppl 2):A-298.
5. AMITIZA package insert. Bethesda, MD: Sucampo Pharmaceuticals, Inc.; 2008.